CheckMate 384: Phase 3b/4 Trial of Nivolumab 480 mg Q4W vs 240 mg Q2W After ≤ 12 Months of Nivolumab in Previously Treated Advanced Non-Small Cell Lung Cancer

Edward B. Garon,[1] Niels Reinmuth,[2] Lionel Falchero,[3] Yolanda García,[4] José Hureaux,[5] Ira Gore,[6] Ronald P. Harris,[7] Paolo Bidoli,[8] Editta Baldini,[9] Silverio Ros,[10] Eckart Laack,[11] Paul Mitchell,[12] Martin Wolf,[13] Kenneth O'Byrne,[14] Labib Zibdawi,[15] Kevin Jao,[16] David R. Spigel,[17] Ang Li,[18] Sridhar K. Rabindran,[18] Eric Pichon[19]

[1]David Geffen School of Medicine at UCLA/TRIO-US Network, Los Angeles, CA, USA; [2]Asklepios Klinik München-Gauting, Munich, Germany; [3]L'hôpital Nord Ouest, Villefranche-sur-Saône, France; [4]Parc Taulí Hospital Universitari, I3PT, UAB, Sabadell, Spain; [5]CHU Angers, Angers, France; [6]Alabama Oncology, Birmingham, AL, USA; [7]Broome Oncology – USOR, Johnson City, NY, USA; [8]Azienda Socio Sanitaria Territoriale – ASST di Monza, Monza, Italy; [9]Oncology Department, Medical Oncology Division, San Luca Hospital, Lucca, Italy; [10]Department of Internal Medicine, University of Murcia, Murcia, Spain; [11]Hämato-Onkologie Hamburg, Hamburg, Germany; [12]Austin Health, Victoria, Australia; [13]Klinikum Kassel, Hessen, Germany; [14]Princess Alexandra Hospital, Brisbane, Queensland, Australia; [15]Southlake Regional Health Centre, Newmarket, ON, Canada; [16]Hôpital du Sacré-Coeur de Montréal, Montreal, QC, Canada; [17]Sarah Cannon Research Institute/Tennessee Oncology Nashville, PLLC, Nashville, TN, USA; [18]Bristol-Myers Squibb, Princeton, NJ, USA; [19]CHRU Bretonneau, Tours, France

Background

- Nivolumab is approved at a fixed dose of 240 mg every 2 weeks (Q2W) across multiple tumors in several countries globally[1–4]
- Nivolumab is also approved at 480 mg every 4 weeks (Q4W) for multiple tumor types in several countries, including non-small cell lung cancer (NSCLC) in the USA and Canada[3,4]
 - Approvals for 480 mg Q4W dosing were based on pharmacokinetic modeling and limited clinical safety data (n = 61), which predicted that the exposure, safety, and efficacy of this dose would be similar to that of the body weight–based 3 mg/kg Q2W[5,6]
- Decreasing the frequency of nivolumab administration may improve convenience while maintaining efficacy and safety
- Here we present an interim analysis of CheckMate 384, a study evaluating 480 mg Q4W nivolumab dosing in patients with previously treated advanced NSCLC with disease control on nivolumab

Methods

Study design

- CheckMate 384 (NCT02713867) is a phase 3b/4, open-label, randomized study investigating 2 approved doses of nivolumab (480 mg Q4W vs 240 mg Q2W) in patients with previously treated advanced NSCLC following up to 12 months of prior treatment with nivolumab 3 mg/kg or 240 mg, each Q2W (**Figure 1**)
- Patients were required to have 2 consecutive assessments of complete response (CR), partial response (PR), or stable disease (SD) on prior nivolumab treatment
- Patients were stratified by tumor histology (squamous vs non-squamous) and response to prior nivolumab therapy at randomization (CR/PR vs SD)

Assessments

- Tumor response: assessed using RECIST v1.1[7] every 8 weeks for the first year after randomization, then every 3 months for the second year
- Safety: adverse events (AEs) were assessed continuously through 100 days after the last dose, and graded using National Cancer Institute Common Terminology Criteria for Adverse Events (CTCAE) v4.0

Figure 1. CheckMate 384 study design

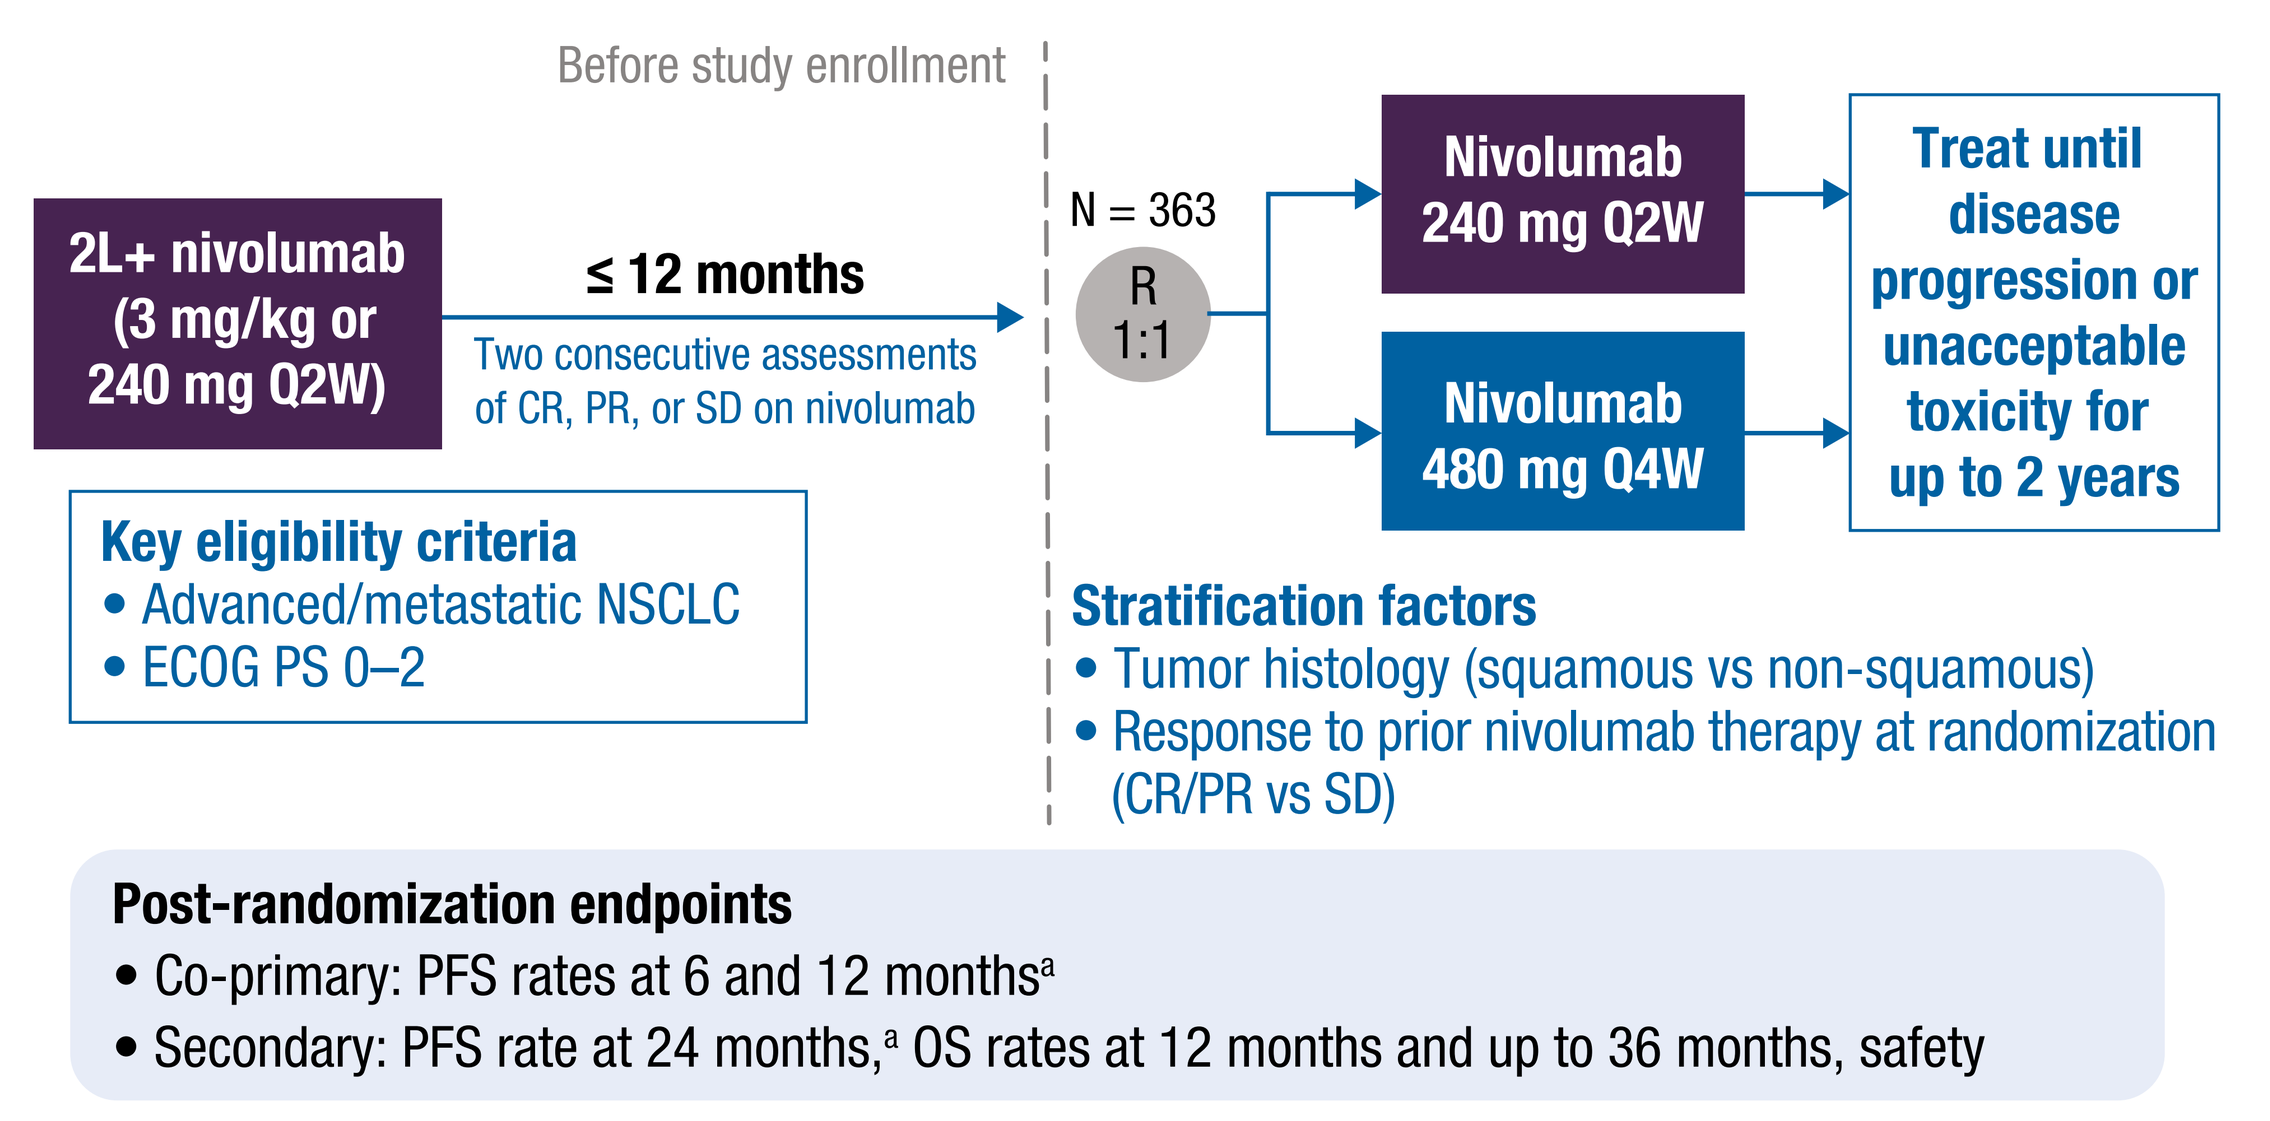
Before study enrollment
2L+ nivolumab (3 mg/kg or 240 mg Q2W)
≤ 12 months
Two consecutive assessments of CR, PR, or SD on nivolumab
N = 363
R 1:1
Nivolumab 240 mg Q2W
Nivolumab 480 mg Q4W
Treat until disease progression or unacceptable toxicity for up to 2 years
Key eligibility criteria
• Advanced/metastatic NSCLC
• ECOG PS 0–2
Stratification factors
• Tumor histology (squamous vs non-squamous)
• Response to prior nivolumab therapy at randomization (CR/PR vs SD)
Post-randomization endpoints
• Co-primary: PFS rates at 6 and 12 months[a]
• Secondary: PFS rate at 24 months,[a] OS rates at 12 months and up to 36 months, safety

Database lock: June 4, 2018.
[a]As measured by investigator-assessed response using RECIST v1.1.
2L+, second or later line; PFS, progression-free survival; PS, performance status; R, randomization.

Statistical considerations

- CheckMate 384 was originally designed as a non-inferiority study (N = 600 planned) to evaluate the efficacy and safety of nivolumab 480 mg Q4W vs 240 mg Q2W with a –10% non-inferiority margin and one-sided 95% CI
- Changes in the treatment landscape of first-line NSCLC have slowed enrollment of second-line immuno-oncology–naive patients with NSCLC into the study. Enrollment was stopped early and the sample size was reduced to N = 363
- With this reduction in sample size, there was insufficient power to conduct the non-inferiority analyses. Therefore, the statistical plan was changed to include descriptive analyses only, with one-sided 95% CI
- Data presented here are from an interim analysis (N = 329). A total of 34 additional patients have been enrolled after this analysis was performed and a final analysis will be conducted when all patients have a minimum of 12 months' follow-up

Results

Patients and disposition

- Patients (N = 329) were randomized at 84 sites in 7 countries (Australia, Canada, France, Germany, Italy, Spain, and the USA)
- Baseline characteristics were generally balanced between treatment arms (**Table 1**)
- At database lock (June 4, 2018), 74 (45%) and 80 (50%) patients in the 480-mg Q4W and 240-mg Q2W arms, respectively, had discontinued treatment; reasons for discontinuations are shown in **Table 2**

Table 1. Baseline characteristics

Characteristic	480 mg Q4W (n = 166)	240 mg Q2W (n = 163)
Age, median (range), years	67 (40–87)	67 (44–85)
Female, n (%)	44 (26)	54 (33)
Weight, median (range), kg	75 (44–133)	73 (34–132)
ECOG PS,[a] n (%)		
0	72 (43)	62 (38)
1	88 (53)	93 (57)
Current or former smoker,[b] n (%)	149 (90)	156 (96)
Tumor histology,[c] n (%)		
Squamous	58 (35)	56 (34)
Non-squamous	108 (65)	107 (66)
Previous lines of systemic therapy (prior to nivolumab),[d] n (%)		
1	98 (59)	115 (71)
2+	66 (40)	46 (28)
Duration of nivolumab treatment prior to randomization,[e] n (%)		
Median (range), months	5.2 (2.4–24.0)	5.6 (1.8–12.7)
3 – < 6 months	95 (57)	81 (50)
6 – < 9 months	37 (22)	42 (26)
9 – < 12 months	26 (16)	29 (18)
Ongoing response to nivolumab at randomization,[c] n (%)		
CR/PR	59 (36)	58 (36)
SD	107 (64)	105 (64)

[a]Not reported in 3 (2%) and 3 (2%) patients in the 480-mg Q4W and 240-mg Q2W arms, respectively; 3 (2%) and 5 (3%) patients, respectively, had an ECOG PS of 2. [b]Not reported in 1 (1%) patient in the 480-mg Q4W arm; 16 (10%) and 7 (4%) patients in the 480-mg Q4W and 240-mg Q2W arms, respectively, had never smoked. [c]Interactive Web Response System source. [d]2 (1%) patients in each arm had no prior systemic therapy. [e]Not reported in 1 (1%) patient in the 480-mg Q4W arm; duration was < 3 months for 5 (3%) and 8 (5%) patients in the 480-mg Q4W and 240-mg Q2W arms, respectively, and ≥ 12 months for 2 (1%) and 3 (2%) patients, respectively (patients were permitted to continue to receive pre-study nivolumab treatment during screening assessments).

Table 2. Post-randomization patient disposition

	480 mg Q4W (n = 166)	240 mg Q2W (n = 163)
Treated, n (%)	164 (99)	161 (99)
Still on therapy, n (%)[a]	90 (55)	81 (50)
Discontinued treatment, n (%)[a]	74 (45)	80 (50)
Disease progression	44 (27)	47 (29)
Study drug toxicity	12 (7)	14 (9)
Death	5 (3)	3 (2)
AE unrelated to study drug	3 (2)	4 (2)
Patient request to discontinue study treatment	5 (3)	3 (2)
Patient withdrew consent	1 (1)	2 (1)
Maximum clinical benefit	0	1 (1)
Patient no longer meets study criteria	1 (1)	1 (1)
Poor/non-compliance	1 (1)	0
Other	2 (1)	5 (3)

[a]Percentages are based on all treated patients.

- Median follow-up was 9.4 months (480 mg Q4W) and 10.2 months (240 mg Q2W), with a minimum follow-up of 2.2 months
- Median duration (between first dose and treatment discontinuation/cut-off) of post-randomization study therapy was 7.5 months (480 mg Q4W) and 7.1 months (240 mg Q2W)

Efficacy

- Post-randomization PFS rates at 6 months appeared similar between treatment arms (**Figure 2**, **Table 3**)
 - Median PFS was 12.1 months (480 mg Q4W) vs 12.2 months (240 mg Q2W); hazard ratio: 0.96 (one-sided 95% CI, not applicable [NA]–1.29)
- Post-randomization PFS rates at 6 months in pre-defined subgroups seemed similar between treatment arms (**Table 3**)

Figure 2. Post-randomization PFS with nivolumab Q4W vs Q2W[a]

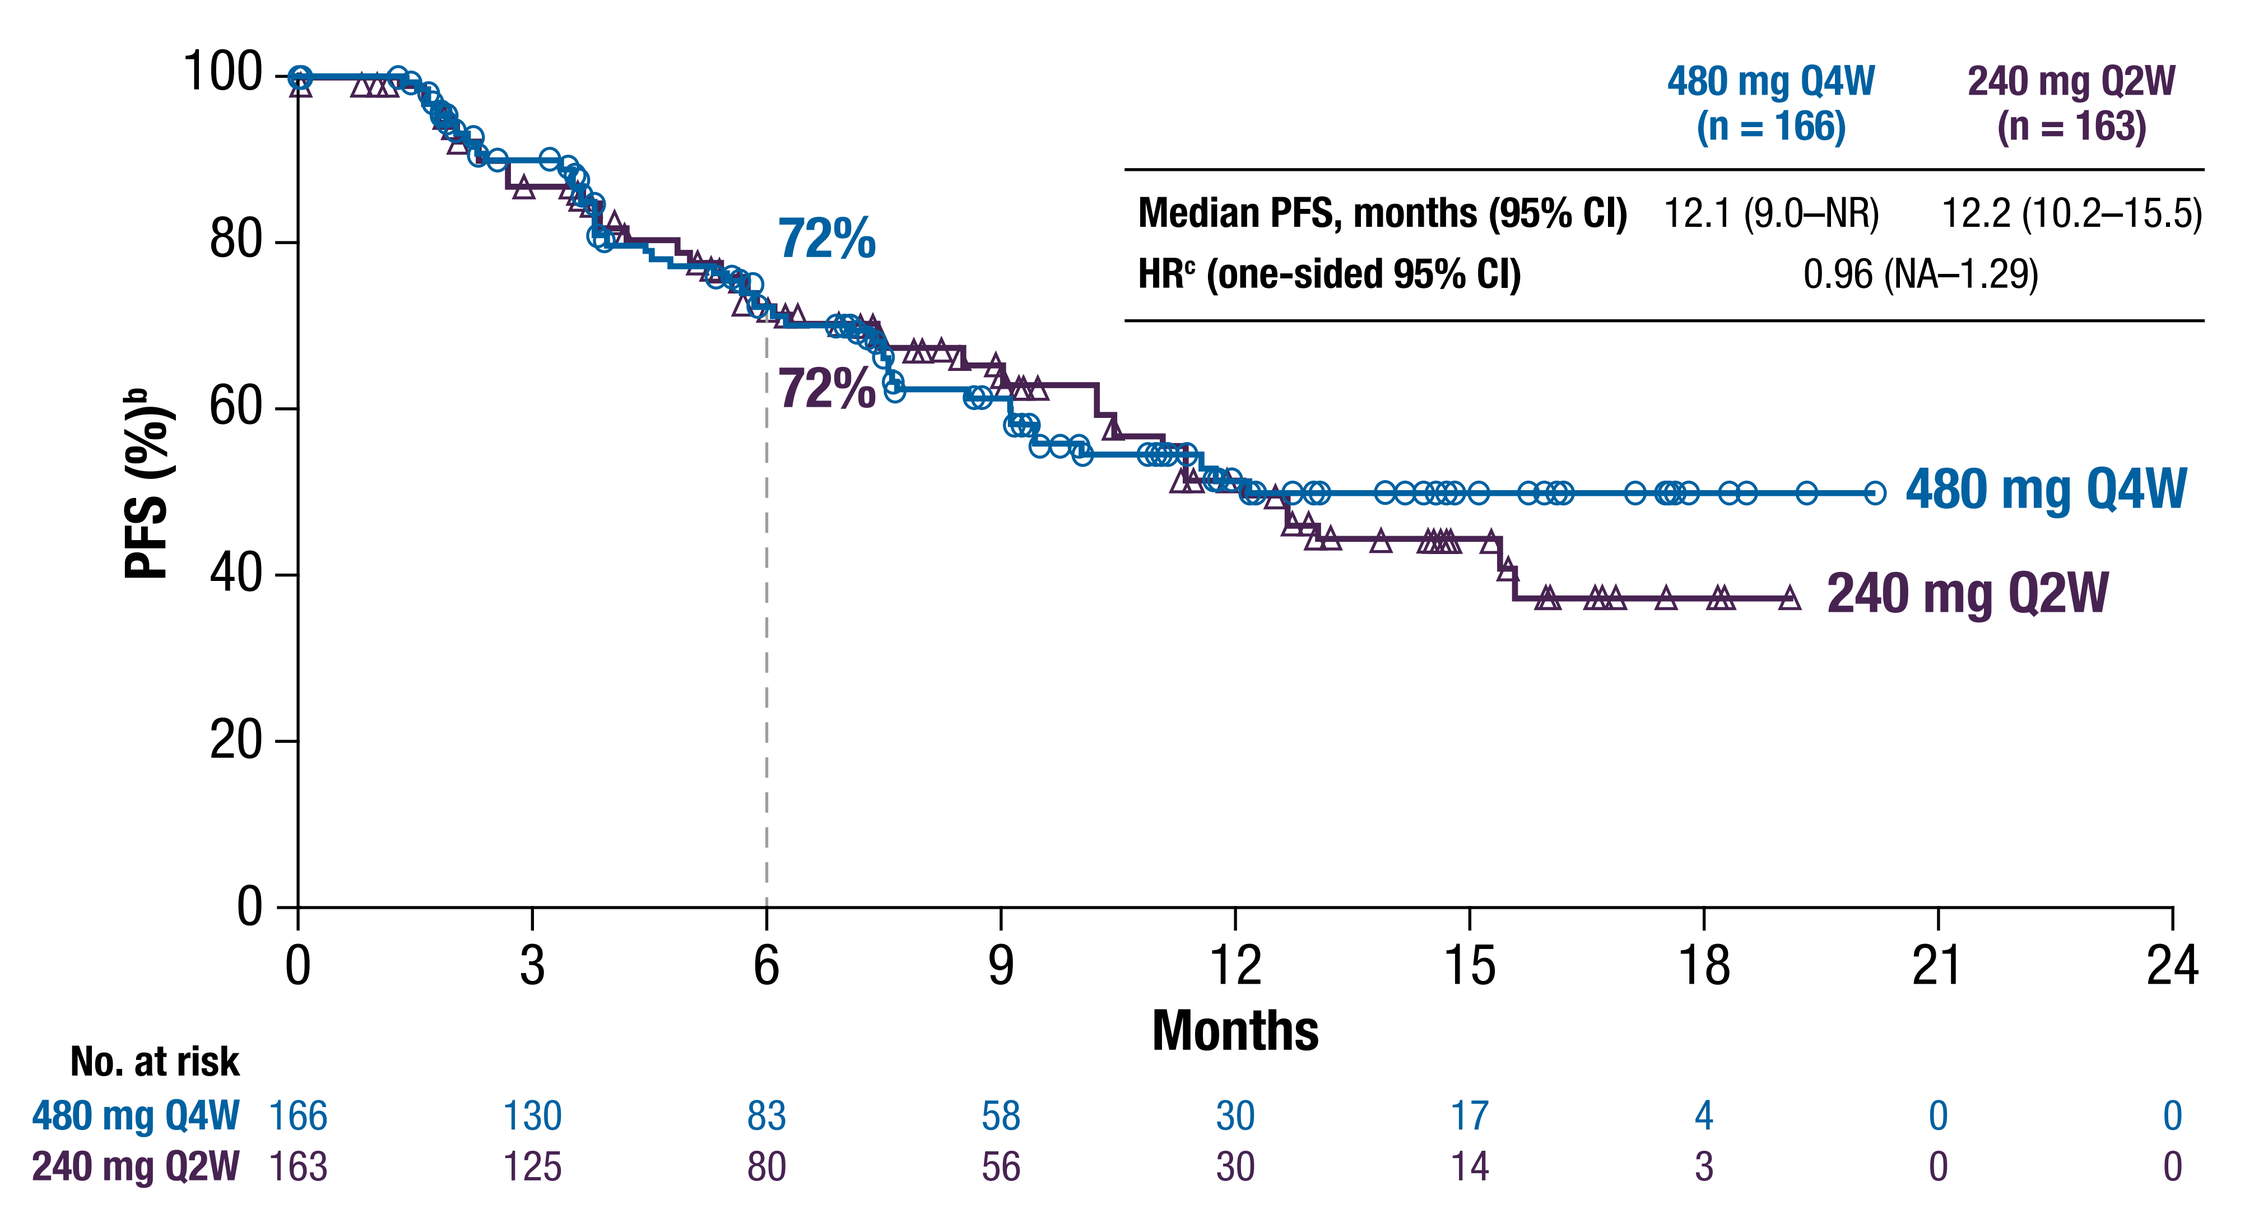
480 mg Q4W (n = 166)
240 mg Q2W (n = 163)
Median PFS, months (95% CI) 12.1 (9.0–NR) 12.2 (10.2–15.5)
HR[c] (one-sided 95% CI) 0.96 (NA–1.29)
72%
72%
480 mg Q4W
240 mg Q2W
PFS (%)[b]
100
80
60
40
20
0
0 3 6 9 12 15 18 21 24
Months
No. at risk
480 mg Q4W 166 130 83 58 30 17 4 0 0
240 mg Q2W 163 125 80 56 30 14 3 0 0

PFS measured by investigator-assessed response using RECIST v1.1; ~80% and ~60% of patients reached 6 months and 12 months of follow-up, respectively. [a]Interim analysis. [b]Unstratified. [c]Adjusted for stratification factors: tumor histology (squamous vs non-squamous) and response category at randomization (CR/PR vs SD). NR, not reached.

Table 3. Post-randomization 6-month PFS rates overall and by subgroup[a]

	480 mg Q4W 6-month PFS rate, %	240 mg Q2W 6-month PFS rate, %	Difference of 6-month PFS rates, % (one-sided 95% CI)[b]
Overall (N = 329)			
Stratified	75	80	–1.9 (–10.0, NA)[c]
Unstratified	72	72	–0.4 (–9.4, NA)
Age			
< 65 years (n = 130)	80	76	4.1 (–9.3, NA)
≥ 65 years (n = 199)	67	70	–3.4 (–15.2, NA)
Sex			
Male (n = 231)	70	72	–2.2 (–13.2, NA)
Female (n = 98)	76	72	4.1 (–11.6, NA)
Baseline ECOG PS			
0 (n = 134)	80	75	4.4 (–8.4, NA)
1 (n = 181)	65	68	–2.8 (–15.7, NA)
Histology			
Squamous (n = 114)	68	64	3.2 (–12.8, NA)
Non-squamous (n = 215)	74	76	–2.1 (–12.8, NA)
Lines of prior therapy			
1 (n = 213)	69	76	–7.0 (–18.2, NA)
2+ (n = 112)	76	61	14.5 (–1.5, NA)
Duration of prior nivolumab treatment			
3 – < 6 months (n = 176)	67	71	–4.5 (–17.2, NA)
6 – < 9 months (n = 79)	71	74	–3.6 (–22.6, NA)
9 – < 12 months (n = 55)	92	77	14.7 (–1.7, NA)
Response at randomization			
CR/PR (n = 117)	84	88	–3.5 (–15.2, NA)
SD (n = 212)	65	64	0.6 (–11.3, NA)

[a]Unstratified PFS rates from the randomized population unless otherwise noted; only groups with ≥ 10 patients per arm are included. [b]The difference of PFS rates is 480 mg Q4W minus 240 mg Q2W. [c]Adjusted for stratification factors: tumor histology (squamous vs non-squamous) and response category at randomization (CR/PR vs SD) and calculated using inverse variance as weights.

Safety

- Overall post-randomization safety profiles appeared comparable between treatment arms (**Table 4**, **Figure 3**)
 - Treatment-related AEs (TRAEs) of any grade were reported in 48% vs 61% of patients receiving 480 mg Q4W vs 240 mg Q2W nivolumab, respectively (**Table 4**)
 - TRAEs leading to discontinuations were reported in 6% vs 9% of patients, respectively (**Table 4**)
 - In both treatment arms, the most common categories of treatment-related select AEs were skin, endocrine, and gastrointestinal; rates of select grade 3–4 AEs were low across categories (**Figure 3**)
- No patients died due to study drug toxicity in either treatment arm

Table 4. Safety summary of post-randomization TRAEs

TRAE,[a] n (%)	480 mg Q4W (n = 164[b]) Any grade	480 mg Q4W (n = 164[b]) Grade 3–4	240 mg Q2W (n = 161[b]) Any grade	240 mg Q2W (n = 161[b]) Grade 3–4
Any TRAE	79 (48)	14 (8)	98 (61)	20 (12)
Serious TRAEs	7 (4)	6 (4)	9 (6)	8 (5)
TRAEs leading to discontinuation	10 (6)	3 (2)	14 (9)	8 (5)
Most frequent TRAEs (≥ 5%[c])				
Diarrhea	18 (11)	3 (2)	13 (8)	1 (1)
Hypothyroidism	14 (8)	0	11 (7)	0
Fatigue	13 (8)	1 (1)	21 (13)	1 (1)
Asthenia	8 (5)	0	9 (6)	1 (1)
Pruritus	7 (4)	0	14 (9)	1 (1)
Lipase increased	5 (3)	2 (1)	9 (6)	6 (4)
Dry skin	2 (1)	0	9 (6)	0
Treatment-related deaths	0		0	

Similar safety was observed across subgroups by weight; few patients were represented in the < 50-kg and ≥ 110-kg subgroups. [a]Includes events reported between first dose and 30 days after last dose of study therapy. [b]Treated population. [c]In either group.

Figure 3. Summary of post-randomization treatment-related select AEs[a]

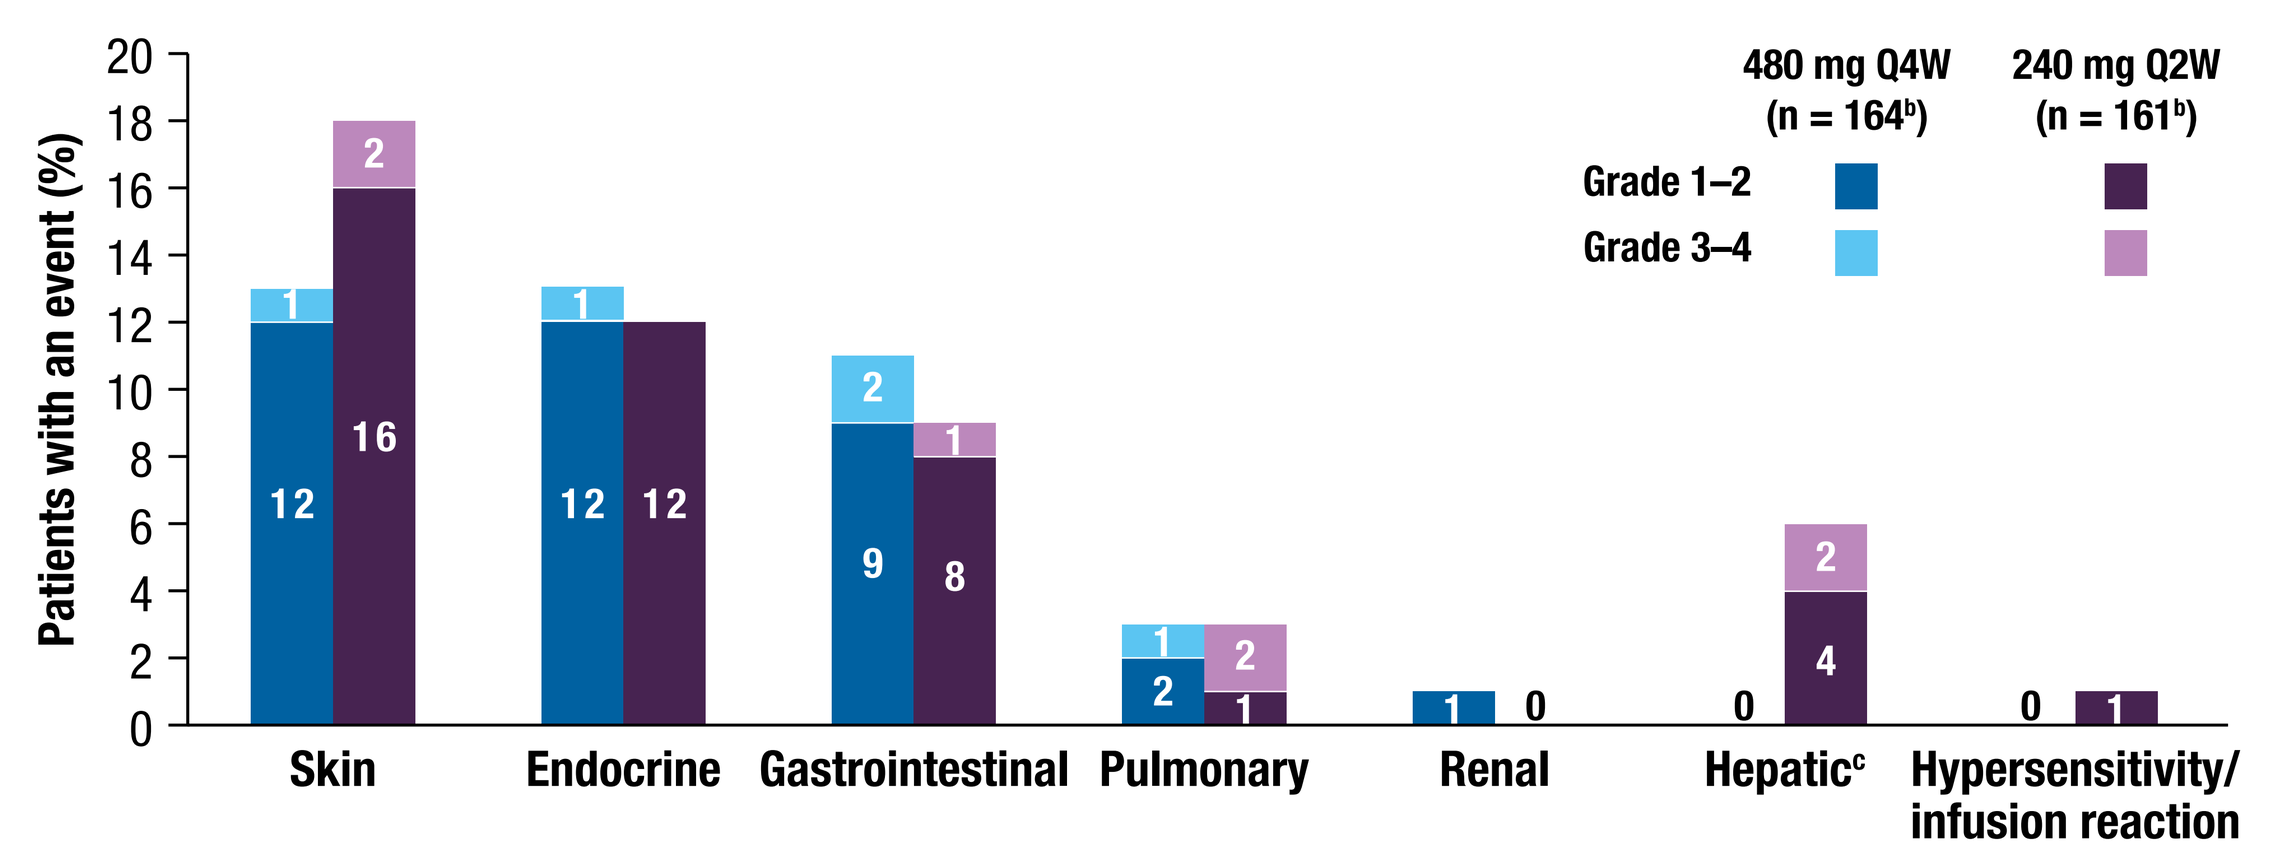
480 mg Q4W (n = 164[b])
240 mg Q2W (n = 161[b])
Grade 1–2
Grade 3–4
Patients with an event (%)
20
18
16
14
12
10
8
6
4
2
0
12
1
16
2
12
1
12
9
2
8
1
2
1
1
2
1
0
0
4
2
0
1
Skin
Endocrine
Gastrointestinal
Pulmonary
Renal
Hepatic[c]
Hypersensitivity/ infusion reaction

[a]Includes events reported between first dose and 30 days after last dose of study therapy; select AEs are those with potential immunologic etiology that require frequent monitoring/intervention. [b]Treated population. [c]Differences in drug-related hepatic select AEs between treatment arms are likely due to variations in the coding for AEs; results of liver function tests were similar between the two arms.

Conclusions

- In this descriptive analysis, nivolumab 480 mg Q4W appeared to show similar efficacy and safety to 240 mg Q2W in patients with advanced NSCLC following disease control with prior nivolumab
 - Post-randomization PFS rates at 6 months appeared similar in both treatment arms
 - No new safety signals were observed with either dose, and trends in TRAEs seemed comparable in both arms
- This study offers support for the use of nivolumab Q4W dosing as a more convenient option for patients with advanced NSCLC with response or stable disease on nivolumab (≤ 12 months)
- Overall, these clinical data are in agreement with pharmacokinetic modeling and provide further evidence supporting a 480-mg Q4W nivolumab dosing regimen

References

1. Opdivo (nivolumab) [summary of product characteristics]. Uxbridge, UK: Bristol-Myers Squibb Company; January 2019.
2. ONO Pharmaceutical Co., Ltd. press release. https://www.ono.co.jp/eng/news/pdf/sm_cn180821.pdf. Published August 21, 2018. Accessed January 9, 2019.
3. OPDIVO® (nivolumab) [package insert]. Princeton, NJ: Bristol-Myers Squibb Company; February 2019.
4. OPDIVO® (nivolumab) [Canadian product monograph]. Bristol-Myers Squibb Canada; March 2019.
5. Zhao X, et al. Poster presentation at the AACR Annual Meeting; April 1–5, 2017; Washington, DC, USA. Abstract CT101.
6. Long GV, et al. *Ann Oncol* 2018;29:2208–2213.
7. Eisenhauer EA, et al. *Eur J Cancer* 2009;45:228–247.

Acknowledgments

- The patients and families who made this study possible
- The clinical study teams who participated in the study
- The protocol manager for this study, Tanisha Bryant (Bristol-Myers Squibb)
- Bristol-Myers Squibb (Princeton, NJ) and ONO Pharmaceutical Company Ltd. (Osaka, Japan)
- The study was supported by Bristol-Myers Squibb
- All authors contributed to and approved the presentation; writing and editorial assistance was provided by Namiko Abe, PhD, of Caudex, and Leah Bernstein, PhD, of Evidence Scientific Solutions, Inc, funded by Bristol-Myers Squibb

Scientific Content On-demand

To request a copy of this poster:

Scan QR code via a barcode reader application

By requesting this content, you agree to receive a one-time communication using automated technology. Msg. & data rates may apply. Links are valid for 30 days after the congress presentation date.

Presented at the 2019 Florida Society of Clinical Oncology (FLASCO) Spring Session; April 11–13, 2019; Kissimmee, FL, USA

Reused with permission from the American Society of Clinical Oncology (ASCO). This abstract was accepted and previously presented at the 2019 ASCO-SITC Clinical Immuno-Oncology Symposium. All rights reserved.

Email: EGaron@mednet.ucla.edu

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the authors of this poster