Effect of dose adjustments on overall survival in patients with metastatic pancreatic ductal adenocarcinoma treated with NALIRIFOX: a *post hoc* analysis of NAPOLI 3

Anjan Patel,[1] Ashley Laursen,[2] Paul Cockrum,[2] Yutong Liu,[3] Andy Surinach,[3] Whitney Rhodes,[3] Li Zhang,[2] Jia Li,[2] Fiona Maxwell,[4] Zev A Wainberg,[5] Eileen M O'Reilly[6]

[1]Florida Cancer Specialists & Research Institute, Sarasota, FL, USA; [2]Ipsen, Cambridge, MA, USA; [3]Genesis Research, Hoboken, NJ, USA; [4]Ipsen, London, UK; [5]University of California Los Angeles, Los Angeles, CA, USA; [6]Memorial Sloan Kettering Cancer Center, New York, NY, USA

The authors would like to thank Dr Pareshkumar Patel for presenting this poster at the 2025 FLASCO Spring Congress

To download the poster, please scan the QR code.

Copies of this poster obtained through the Quick Response (QR) code are for personal use only and may not be reproduced without permission from the author of this poster.

For further information, please send your question(s) to: **Anjan Patel** (ajpatel@flcancer.com).

KEY LEARNINGS

These data indicate that, in patients who received NALIRIFOX in the NAPOLI 3 trial, dose reductions of liposomal irinotecan or oxaliplatin did not negatively impact OS. This finding supports the potential for treatment optimization in patients who receive first-line NALIRIFOX for the treatment of mPDAC.

BACKGROUND

- NALIRIFOX (liposomal irinotecan in combination with 5-fluorouracil/leucovorin plus oxaliplatin) is an FDA-approved, EMA-approved, and NCCN Category 1-recommended option for the first-line treatment of metastatic pancreatic ductal adenocarcinoma (mPDAC).[1–3]
- Approval was based on the results of the NAPOLI 3 trial (NCT04083235; N = 770), in which NALIRIFOX significantly improved overall survival (OS; 11.1 vs 9.2 months) and progression-free survival (7.4 vs 5.6 months) in patients with previously untreated mPDAC compared with nab-paclitaxel plus gemcitabine (Gem+NabP).[4]
- In the NAPOLI 3 trial, related treatment-emergent adverse events led to a dose reduction in 198 patients (54%) who received NALIRIFOX and 184 patients (49%) who received Gem+NabP.[4]
- The impact of NALIRIFOX dose reductions on OS has not previously been reported.

AIM

- To examine the impact of liposomal irinotecan and oxaliplatin dose reductions on OS in NAPOLI 3.

METHODS

Study design and patients

- NAPOLI 3 was an open-label, randomized, phase 3 trial conducted at 187 sites in 18 countries worldwide (Figure 1).[4]
- The exploratory endpoint for this *post hoc* analysis was OS in patients with and without dose reduction of liposomal irinotecan or oxaliplatin.

Statistical analysis

- OS was defined as time from randomization to death or censoring date and was evaluated using the Kaplan–Meier method; no statistical tests were performed.
- Dose reductions were recorded for the safety population and were analyzed descriptively.

Figure 1. NAPOLI 3 study design

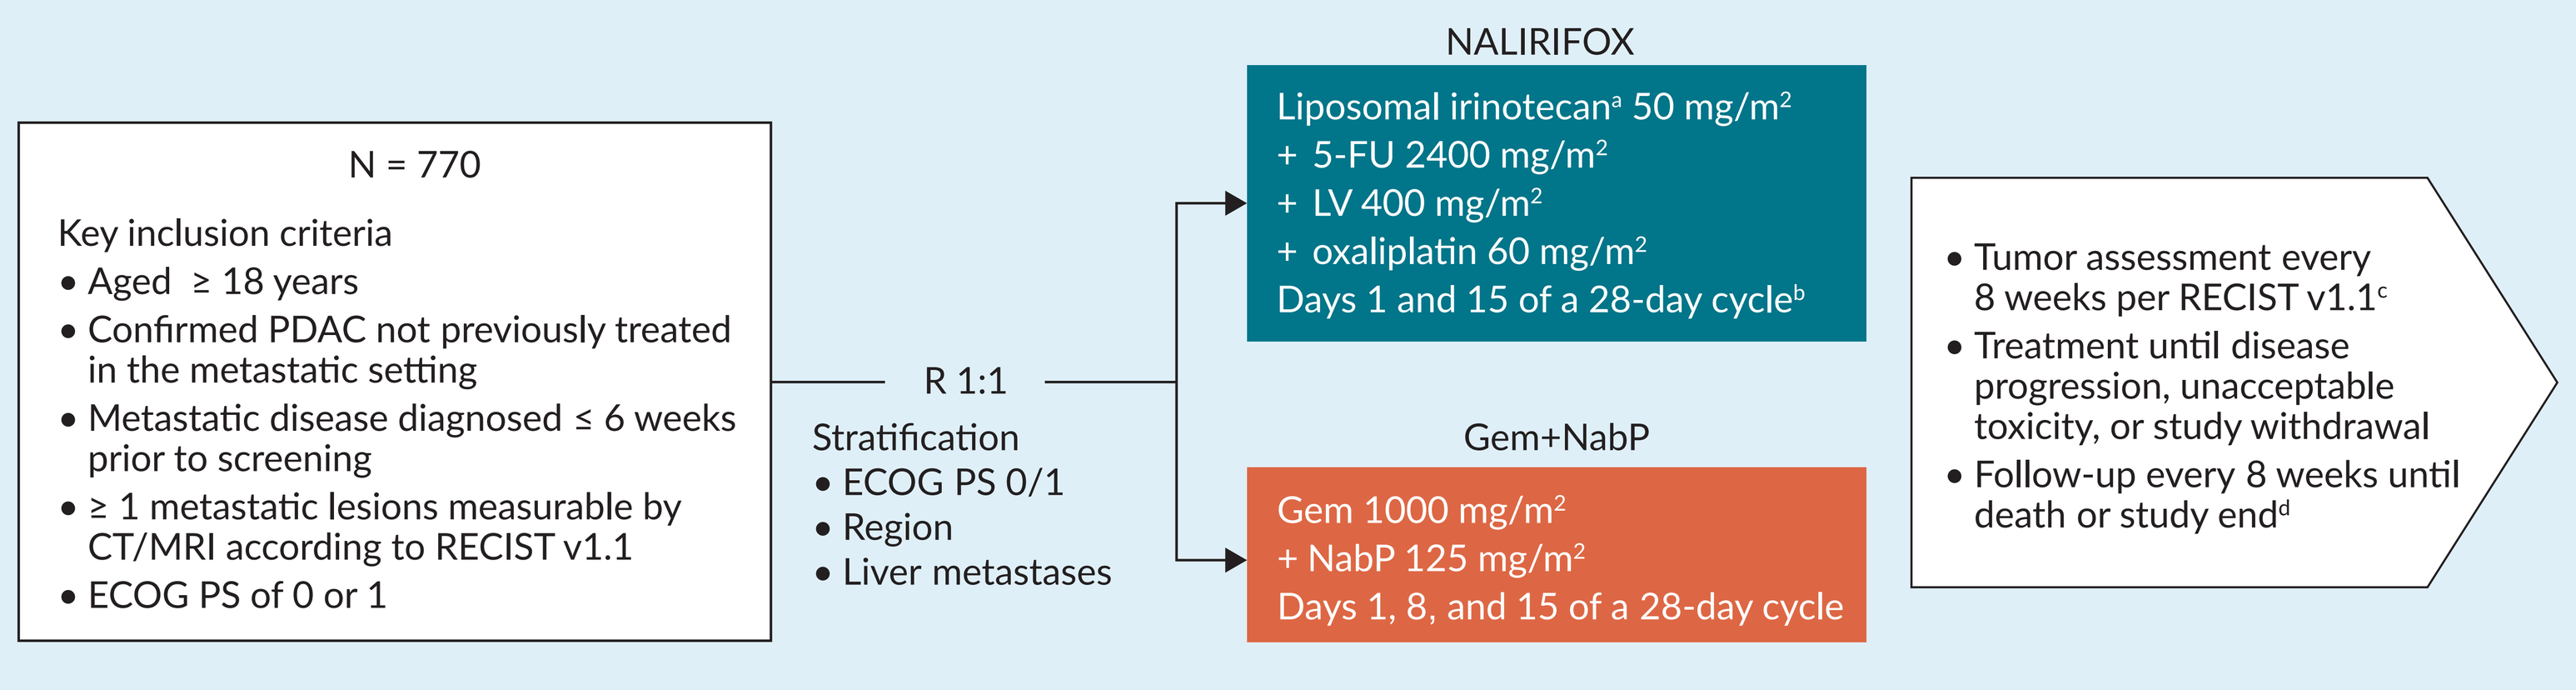
N = 770
Key inclusion criteria
• Aged ≥ 18 years
• Confirmed PDAC not previously treated in the metastatic setting
• Metastatic disease diagnosed ≤ 6 weeks prior to screening
• ≥ 1 metastatic lesions measurable by CT/MRI according to RECIST v1.1
• ECOG PS of 0 or 1
R 1:1
Stratification
• ECOG PS 0/1
• Region
• Liver metastases
NALIRIFOX
Liposomal irinotecan[a] 50 mg/m²
+ 5-FU 2400 mg/m²
+ LV 400 mg/m²
+ oxaliplatin 60 mg/m²
Days 1 and 15 of a 28-day cycle[b]
Gem+NabP
Gem 1000 mg/m²
+ NabP 125 mg/m²
Days 1, 8, and 15 of a 28-day cycle
• Tumor assessment every 8 weeks per RECIST v1.1[c]
• Treatment until disease progression, unacceptable toxicity, or study withdrawal
• Follow-up every 8 weeks until death or study end[d]

[a]Dose expressed as irinotecan free base equivalent. [b]Administered sequentially as a continuous infusion over 46 hours (dose delays and oxaliplatin discontinuation were permitted). [c]Until progressive disease. [d]The study was complete once all patients had discontinued the study treatment and ≥ 543 OS events had occurred.
5-FU, 5-fluorouracil; CT, computed tomography; ECOG PS, Eastern Cooperative Oncology Group performance status; Gem, gemcitabine; LV, leucovorin; MRI, magnetic resonance imaging; NabP, nab-paclitaxel; OS, overall survival; PDAC, pancreatic ductal adenocarcinoma; R, randomization; RECIST, Response Evaluation Criteria in Solid Tumors.

CONCLUSIONS

- The results of this *post hoc* analysis suggest that liposomal irinotecan or oxaliplatin dose reductions do not adversely affect OS.
- Patients with dose reductions of liposomal irinotecan and oxaliplatin had a longer OS, duration of exposure, and higher cumulative dose compared with those who did not.
 - Longer OS may be related to longer time on therapy and an increased likelihood of dose adjustment.
- These data suggest a path forward to further optimize the OS of patients with mPDAC receiving NALIRIFOX.

RESULTS

Patients

- Of the 370 patients who received NALIRIFOX in NAPOLI 3 (safety population), 194 and 217 received dose reductions of liposomal irinotecan and oxaliplatin, respectively (Table 1).

Table 1. Selected demographic and disease characteristics by dose reduction of liposomal irinotecan and oxaliplatin (safety population)

	Overall		North America		Rest of the world	
	Dose not reduced	Dose reduced	Dose not reduced	Dose reduced	Dose not reduced	Dose reduced
Liposomal irinotecan	**n = 176**	**n = 194**	**n = 49**	**n = 63**	**n = 127**	**n = 131**
Age, years, median (range)	64.0 (36.0–81.0)	65.0 (20.0–85.0)	63.0 (43.0–77.0)	66.0 (47.0–85.0)	64.0 (36.0–81.0)	64.0 (20.0–80.0)
Women	70 (39.8)	104 (53.6)	15 (30.6)	32 (50.8)	55 (43.3)	72 (55.0)
ECOG PS score 0	61 (34.7)	94 (48.5)	17 (34.7)	27 (42.9)	44 (34.6)	67 (51.1)
Liver metastases per eCRF	147 (83.5)	148 (76.3)	39 (79.6)	47 (74.6)	108 (85.0)	101 (77.1)
≥ 3 metastatic sites	74 (42.0)	71 (36.6)	23 (46.9)	22 (34.9)	51 (40.2)	49 (37.4)
Oxaliplatin	**n = 153**	**n = 217**	**n = 40**	**n = 72**	**n = 113**	**n = 145**
Age, years, median (range)	64.0 (36.0–81.0)	64.0 (20.0–85.0)	64.0 (43.0–77.0)	65.0 (45.0–85.0)	64.0 (36.0–81.0)	64.0 (20.0–80.0)
Women	58 (37.9)	116 (53.5)	13 (32.5)	34 (47.2)	45 (39.8)	82 (56.6)
ECOG PS score 0	50 (32.7)	105 (48.4)	14 (35.0)	30 (41.7)	36 (31.9)	75 (51.7)
Liver metastases per eCRF	129 (84.3)	166 (76.5)	32 (80.0)	54 (75.0)	97 (85.8)	112 (77.2)
≥ 3 metastatic sites	63 (41.2)	82 (37.8)	17 (42.5)	28 (38.9)	46 (40.7)	54 (37.2)

Data are n (%) unless otherwise stated.
ECOG PS, Eastern Cooperative Oncology Group performance status; eCRF, electronic case report form.

Overall survival

- Patients with dose reductions of liposomal irinotecan or oxaliplatin had longer OS than those without dose reductions; similar results were observed in patients who received treatment in North America and in the rest of the world (Figure 2).
- Median OS was shortest among patients who did not receive a dose reduction of liposomal irinotecan (9.1 months) or oxaliplatin (7.9 months) (Table 2).

Figure 2. Overall survival in patients with and without dose reduction of liposomal irinotecan and oxaliplatin (safety population)

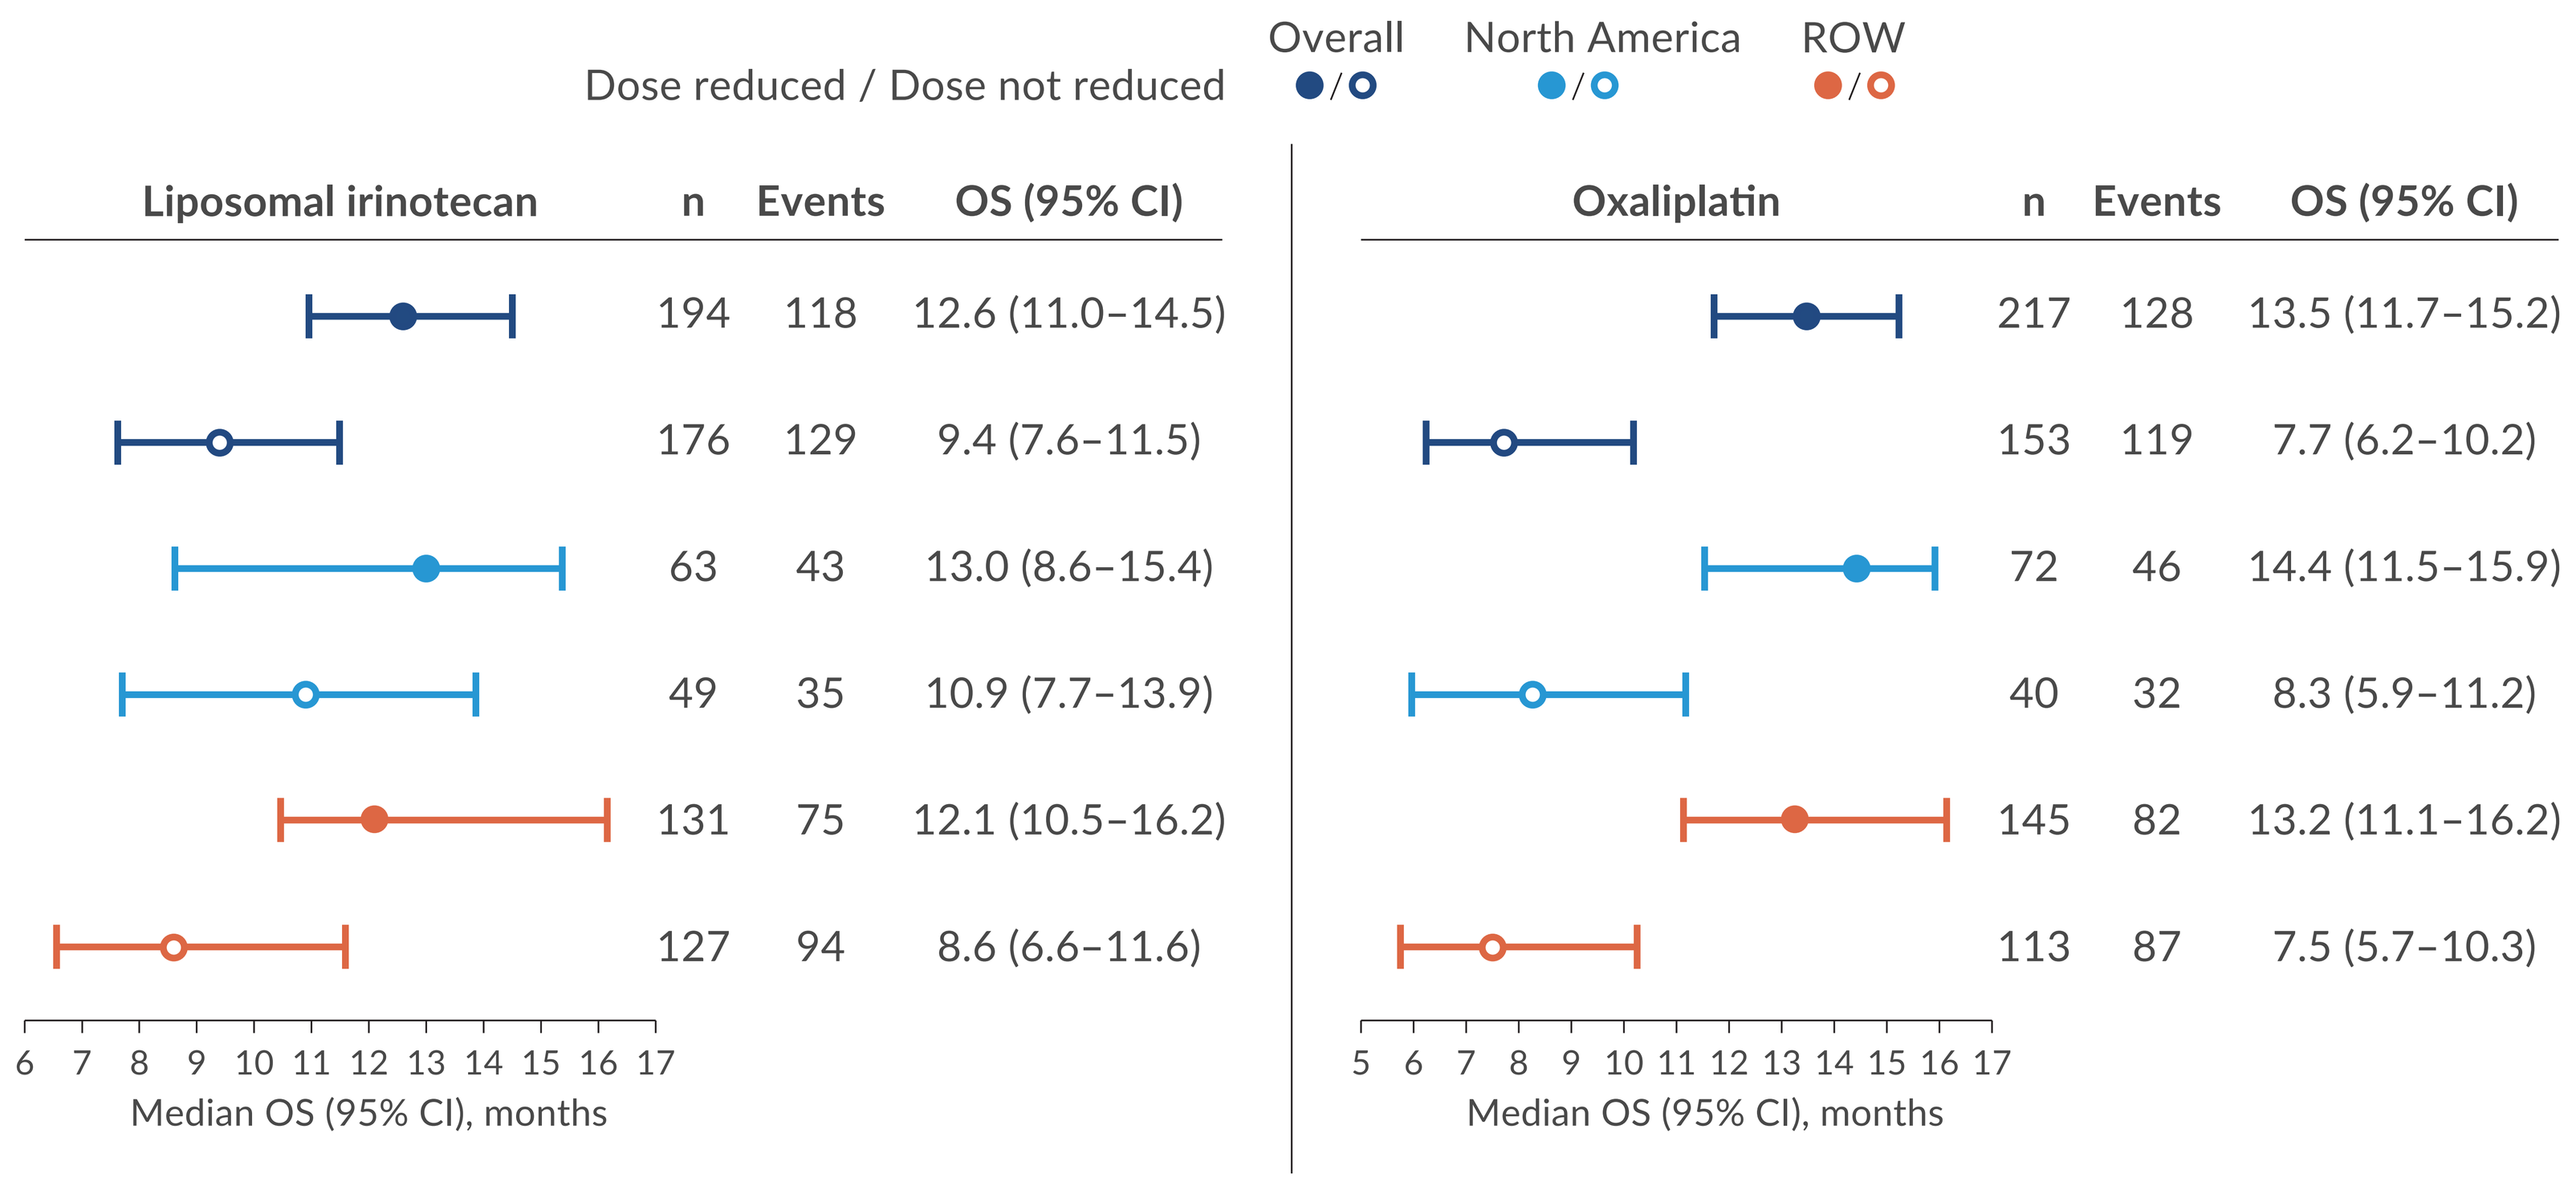
Dose reduced / Dose not reduced
Overall
North America
ROW
Liposomal irinotecan
n
Events
OS (95% CI)
194 118 12.6 (11.0–14.5)
176 129 9.4 (7.6–11.5)
63 43 13.0 (8.6–15.4)
49 35 10.9 (7.7–13.9)
131 75 12.1 (10.5–16.2)
127 94 8.6 (6.6–11.6)
6 7 8 9 10 11 12 13 14 15 16 17
Median OS (95% CI), months
Oxaliplatin
n
Events
OS (95% CI)
217 128 13.5 (11.7–15.2)
153 119 7.7 (6.2–10.2)
72 46 14.4 (11.5–15.9)
40 32 8.3 (5.9–11.2)
145 82 13.2 (11.1–16.2)
113 87 7.5 (5.7–10.3)
5 6 7 8 9 10 11 12 13 14 15 16 17
Median OS (95% CI), months

CI, confidence interval; NALIRIFOX, liposomal irinotecan + 5-fluorouracil/leucovorin + oxaliplatin; OS, overall survival; ROW, rest of world.

Table 2. Median overall survival by lowest dose of liposomal irinotecan and oxaliplatin (safety population)

	Overall survival		
	n	Events	Median (95% CI), months
Lowest dose of liposomal irinotecan			
50 mg/m² (starting dose)	173	129	9.1 (7.5–11.5)
40 mg/m²	118	74	11.8 (10.0–14.4)
32.5 mg/m²	56	30	16.9 (10.4–NE)
25 mg/m²	23	14	13.5 (10.3–NE)
Lowest dose of oxaliplatin			
60 mg/m² (starting dose)	150	117	7.9 (6.3–10.2)
48 mg/m²	116	77	11.7 (9.4–13.9)
39 mg/m²	73	36	17.1 (11.8–NE)
30 mg/m²	30	16	14.4 (12.2–NE)

CI, confidence interval; NE, not estimable.

Treatment exposure and cumulative dose

- Median duration of exposure was longer and cumulative dose was higher among patients who received a dose reduction than among those who did not, for both liposomal irinotecan and oxaliplatin; similar results were observed in patients who received treatment in North America and in the rest of the world (Table 3).

Table 3. Cumulative dose and duration of exposure of liposomal irinotecan and oxaliplatin (safety population)

	Overall		North America		Rest of the world	
	Dose not reduced	Dose reduced	Dose not reduced	Dose reduced	Dose not reduced	Dose reduced
Liposomal irinotecan	**n = 176**	**n = 194**	**n = 49**	**n = 63**	**n = 127**	**n = 131**
Cumulative dose, median (IQR), mg/m²	248.9 (100.0–760.0)	536.0 (295.6–870.1)	403.5 (102.1–809.4)	460.1 (245.9–966.9)	200.9 (99.7–755.0)	544.9 (320.0–824.4)
Duration of exposure at any dose, median (IQR), weeks	10.6 (3.9–35.6)	31.7 (17.1–51.7)	18.1 (4.1–35.0)	25.3 (15.1–53.7)	8.3 (3.0–36.3)	32.1 (18.0–50.1)
Oxaliplatin	**n = 153**	**n = 217**	**n = 40**	**n = 72**	**n = 113**	**n = 145**
Cumulative dose, median (IQR), mg/m²	239.9 (119.1–595.5)	635.6 (360.5–907.3)	327.6 (121.6–628.5)	653.8 (304.9–974.8)	238.1 (60.3–595.4)	635.6 (410.7–860.3)
Duration of exposure at any dose, median (IQR), weeks	8.1 (2.9–23.0)	30.0 (17.3–40.1)	12.1 (3.6–24.7)	25.2 (15.1–43.8)	8.0 (2.1–21.3)	30.1 (18.0–39.7)

IQR, interquartile range.

Reasons for dose reduction and discontinuation

- The most common adverse event (any grade) leading to dose reduction of liposomal irinotecan and oxaliplatin was diarrhea (40% and 36% of patients with dose reductions, respectively).
- The proportion of patients with grade ≥ 3 diarrhea events leading to dose reduction of liposomal irinotecan or oxaliplatin was higher in North America (38% and 35%, respectively) than in the rest of the world (24% and 23%, respectively).
- Among patients who did not receive dose reductions of liposomal irinotecan and oxaliplatin, the most common reasons for treatment discontinuation were disease progression per RECIST (38% and 35%, respectively) and adverse events (19% and 21%, respectively).

Abbreviations EMA, European Medicines Agency; FDA, US Food and Drug Administration; Gem+NabP, nab-paclitaxel plus gemcitabine; mPDAC, metastatic pancreatic ductal adenocarcinoma; NALIRIFOX, liposomal irinotecan + 5-fluorouracil/leucovorin + oxaliplatin; NCCN, National Comprehensive Cancer Network; OS, overall survival; RECIST, Response Evaluation Criteria in Solid Tumors.

Medical writing support The authors thank Liz Sloan, PhD, of Oxford PharmaGenesis, Oxford, UK for providing medical writing and editorial support, which was sponsored by Ipsen in accordance with Good Publication Practice guidelines (GPP 2022).

Author contributions All authors provided substantial contributions to study conception/design or acquisition/analysis/interpretation of data; drafting of the publication or reviewing it critically for important intellectual content; and gave their final approval of the publication.

Disclosures AP: None to disclose.

Presented at the Florida Society of Clinical Oncology (FLASCO) 2025 | April 4–5, 2025 | Orlando, FL, USA.

Previously presented at the American Society of Clinical Oncology Gastrointestinal Cancers Symposium (ASCO GI) 2025 | January 23–25, 2025 | San Francisco, CA, USA.

Acknowledgments The authors thank all patients involved in the study, as well as their caregivers and care teams, and the investigators and research staff in participating institutions.

References

1. US Food and Drug Administration. Prescribing Information: ONIVYDE® (irinotecan liposome injection), for intravenous use. Initial U.S. Approval: 1996. Revised February 2024. Available from: https://d2rkmuse97gwnh.cloudfront.net/a88aa6d6-3ca0-4362-a711-d53c45ae33ff/68b50c8f-8577-4904-950d-d82fa1f91417/68b50c8f-8577-4904-950d-d82fa1f91417_source__v.pdf (Accessed December 2024).
2. European Medicines Agency. Onyvide pegylated liposomal irinotecan hydrochloride trihydrate. Post authorisation summary. Available from: https://www.ema.europa.eu/en/documents/smop/chmp-post-authorisation-summary-positive-opinion-onivyde-pegylated-liposomal-previously-known-onivyde-ii-34_en.pdf (Accessed December 2024).
3. Ipsen. Onivyde® (irinotecan liposome injection). NCCN recommendations. Available from: https://www.onivyde.com/en-us/hcp/nccn-recommendation-irinotecan (Accessed December 2024).
4. Wainberg ZA *et al. Lancet* 2023;402:1272–81.

This study was sponsored by Ipsen.